Costimulatory T cell engagement via a novel bispecific anti-CD137/anti-HER2 protein based on Anticalin® technology

Marlon J. Hinner, Rachida-Siham Bel Aiba, Alexander Wiedenmann, Corinna Schlosser, Andrea Allersdorfer, Gabriele Matschiner, Christine Rothe, Ulrich Moebius, Shane A. Olwill

Pieris Pharmaceuticals, Inc., Lise-Meitner-Str. 30, 85354 Freising, Germany

CRI 2015, New York
September 18th

Abstract

CD137 is a potent costimulatory immunoreceptor and a highly promising target for immunostimulatory cancer therapy. Conventional CD137-targeting antibodies, however, suffer from a lack of tumor-selective activity, which may lead to peripheral toxicity and reduce the available therapeutic window.

To generate a therapeutic that facilitates a CD137-based activation of T cells that is both tumor-target driven and tumor-localized, we generated four variants of a bispecific protein binding to CD137 and the tumor target HER2. The bispecifics are designed to promote CD137 clustering by bridging T cells with HER2-positive tumor cells, and to thereby provide a potent costimulatory signal to tumor antigen-specific T cells.

Here, we describe the generation and characterization of Anticalin-based CD137/HER2 bispecifics. Most importantly, we show *ex vivo* that T cells are efficiently activated when incubated with a bispecific and HER2-positive cells, and that the activation is HER2-dependent. Interestingly, the efficiency of T cell activation differs greatly among constructs of identical target affinity, showing the importance of construct geometry.

Concept: tumor-specific and tumor-localized costimulatory activation of T cells

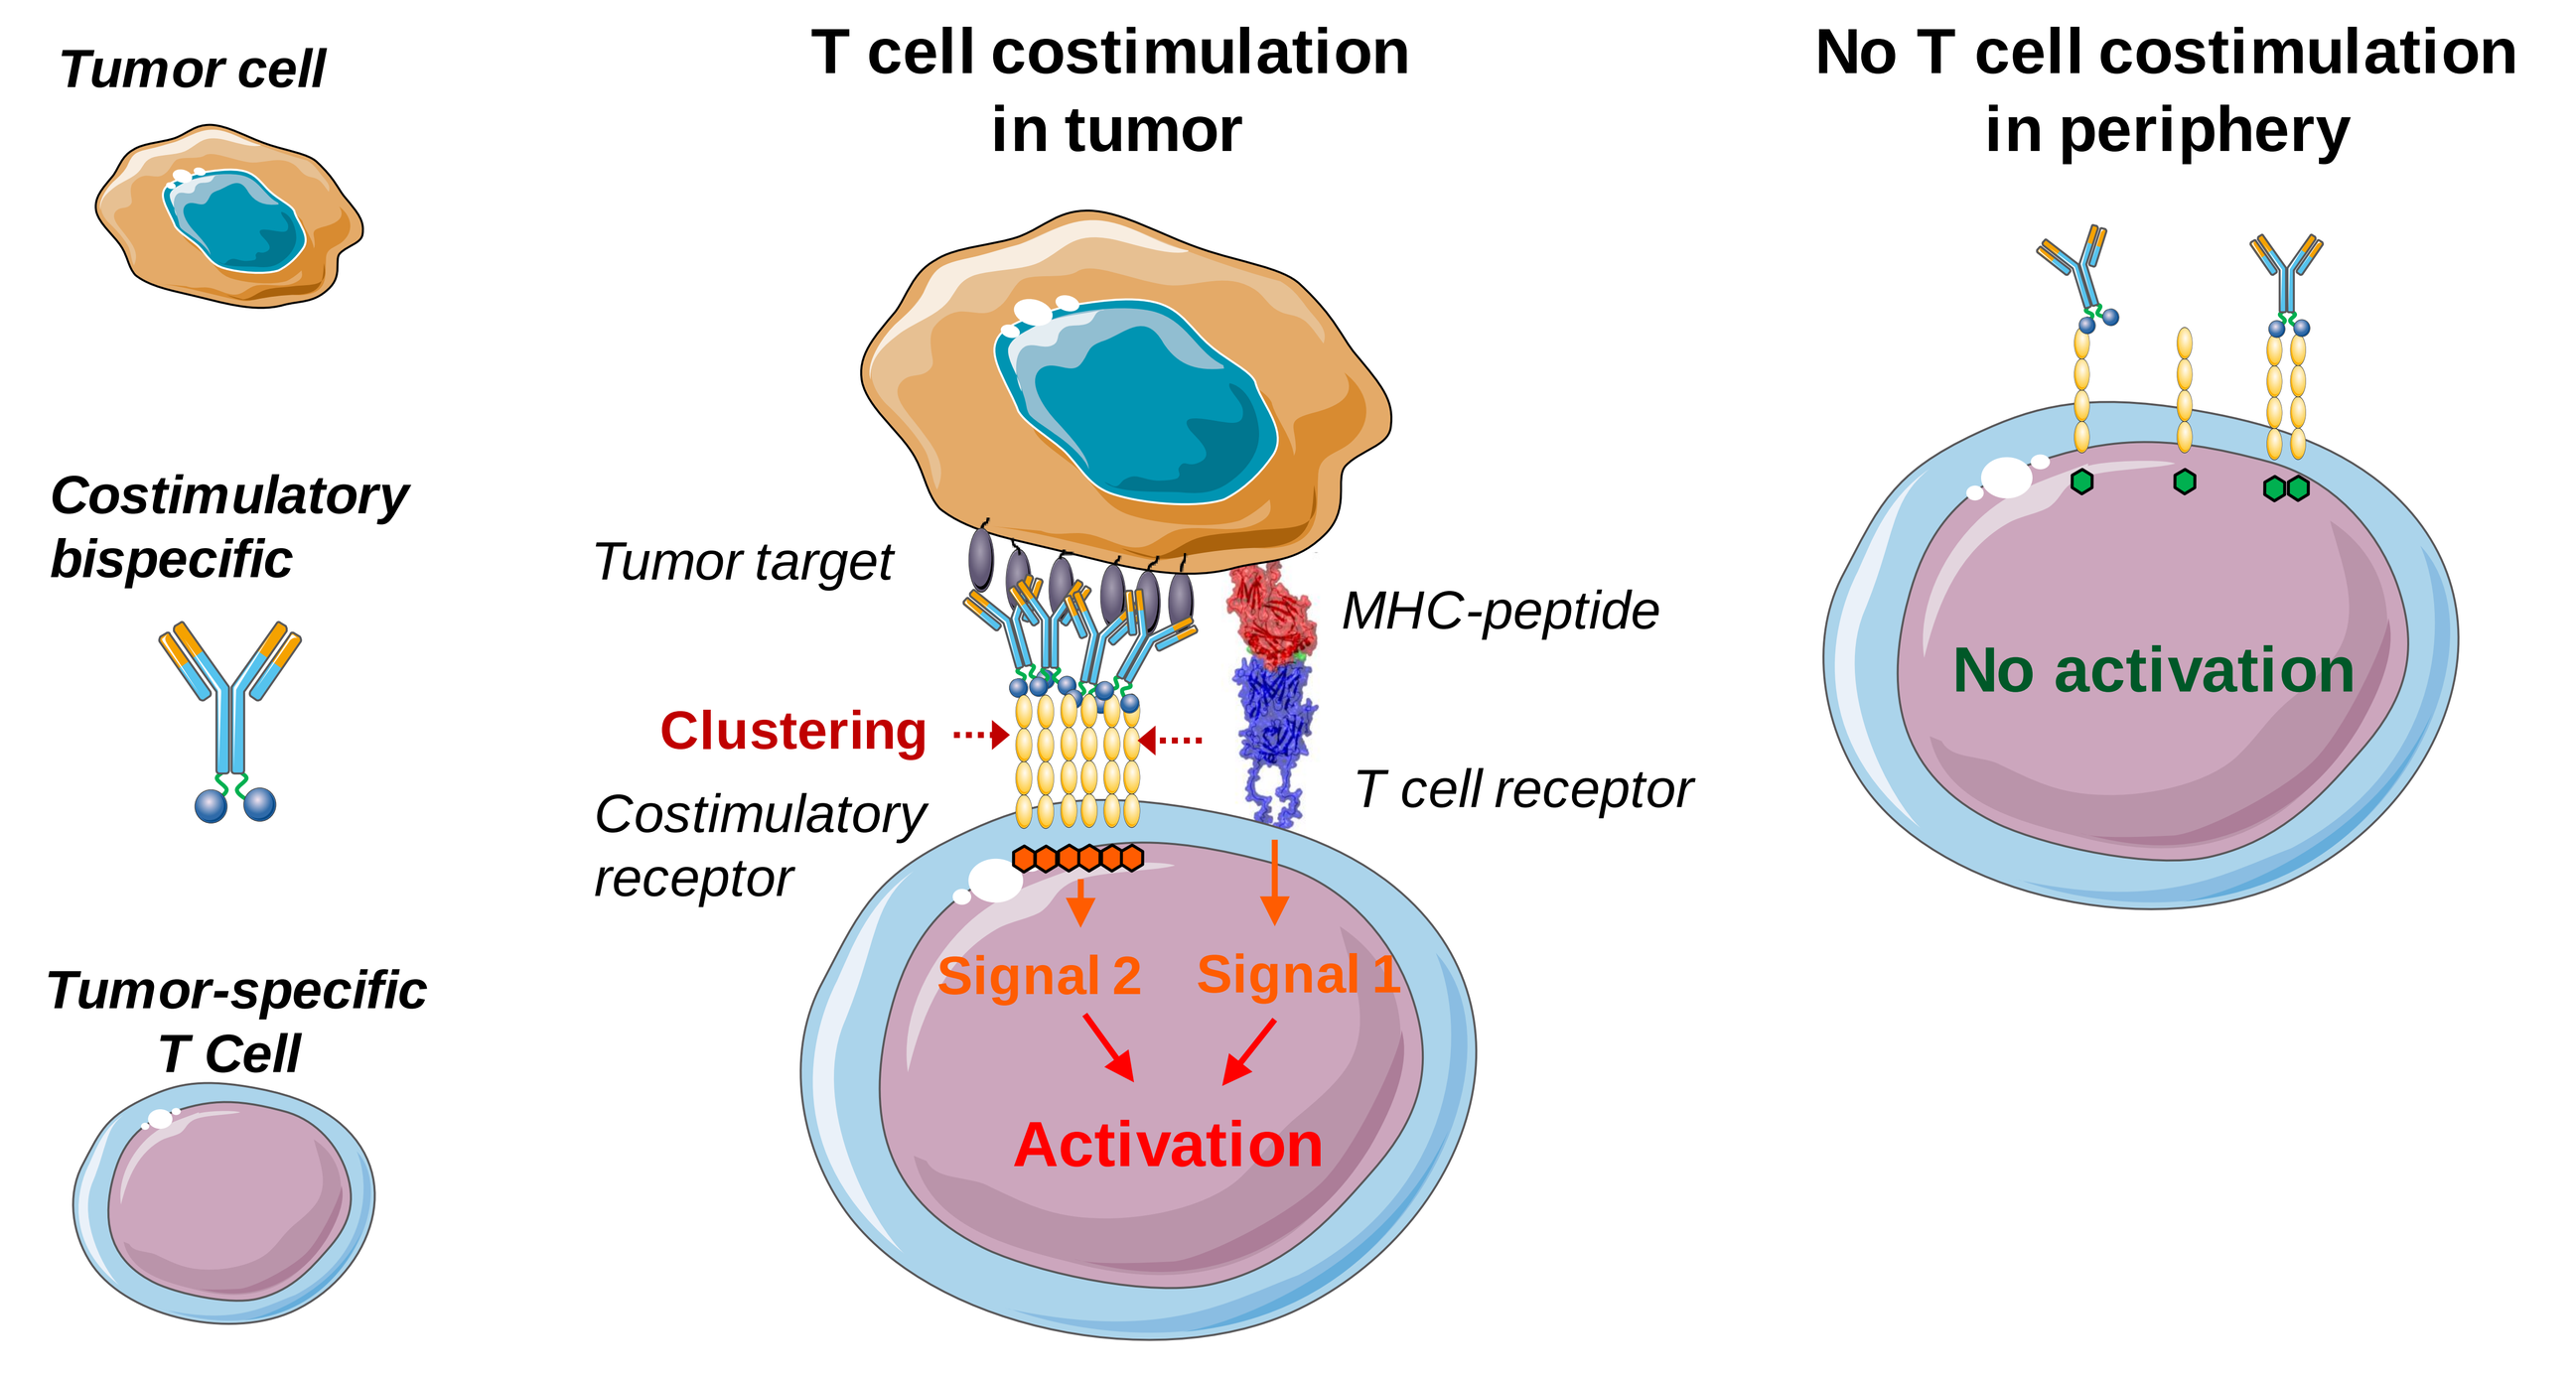
Tumor cell
T cell costimulation in tumor
No T cell costimulation in periphery
Costimulatory bispecific
Tumor target
MHC-peptide
Clustering
Costimulatory receptor
T cell receptor
Tumor-specific T Cell
Signal 2
Signal 1
Activation
No activation

Figure 1. Concept of costimulatory T cell engagement: Within a patient´s tumor, tumor-specific T cells are bridged with tumor cells by a costimulatory bispecific. The resulting clustering of the costimulatory T cell receptor provides a local co-activatory signal to the T cell, further enhancing its T cell receptor (TCR) mediated activity and leading to tumor destruction. Toxic side effects are expected to be manageable, as target-negative cells do not lead to costimulation of T cells due to a lack of target-mediated receptor clustering, and healthy tissue is spared by tumor-costimulated T cells due to the absence of a primary, TCR-mediated signal.

CD137 – the prime costimulatory receptor

Background
- CD137 mainly expressed on activated CD4+ and CD8+ T cells, activated B cells, and natural killer (NK) cells
- CD137 activation occurs via clustering by cell-surface expressed CD137 ligand (mainly on antigen-presenting cells)
- CD137-costimulated T- and NK-cells have enhanced proliferation, proinflammatory cytokine expression and killing capacity

Target validation
- CD137 is a validated marker for tumor-reactive T cells in man[1]
- Anti-CD137 mAbs improve expansion of $CD8^+$ melanoma TIL in adoptive T cell therapy[2]
- CD137 downstream signaling is key to success in clinical CAR-T[3,4]
- CD137 costimulation in NK-cells is currently evaluated in clinical trials[5]
- CD137 expression is localized in the tumor microenvironment[6]
- Costimulatory T cell engagement via CD137 has been demonstrated using aptamer technology[7]

Building block: Anticalin targeting CD137

Discovery
- Phage display of lipocalin library against CD137, followed by affinity maturation

Binding to CD137
- K_D = 2.3nM (SPR)
- EC50(FACS) = 5.9nM
- Non-competitive binding vs CD137L

Biophysical properties
- 100% monomeric expression
- T_M =74°C (DSC)
- Fully stable after 1wk @37°C in PBS, human plasma or mouse plasma

Functional activity
- Ex vivo activation of T cells when coated, no activation when in solution

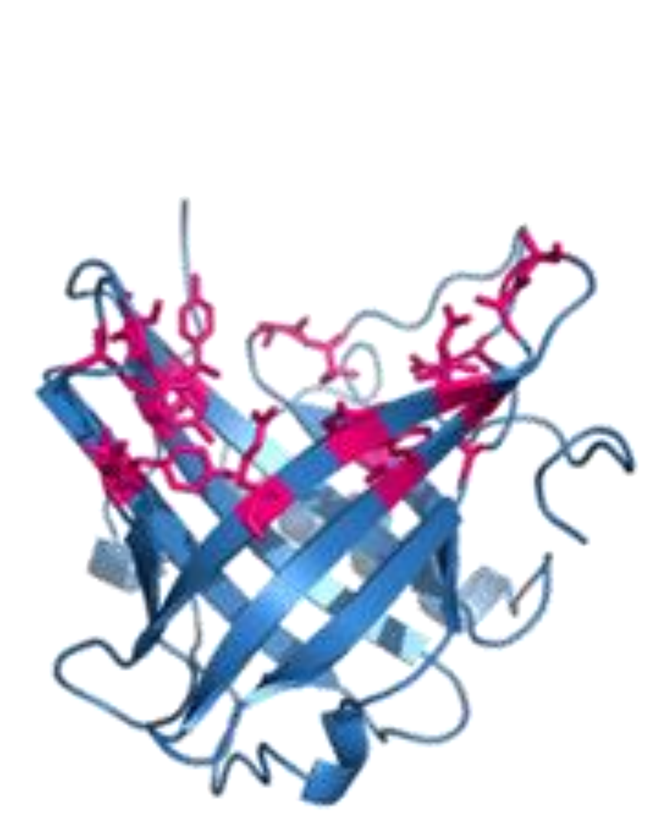

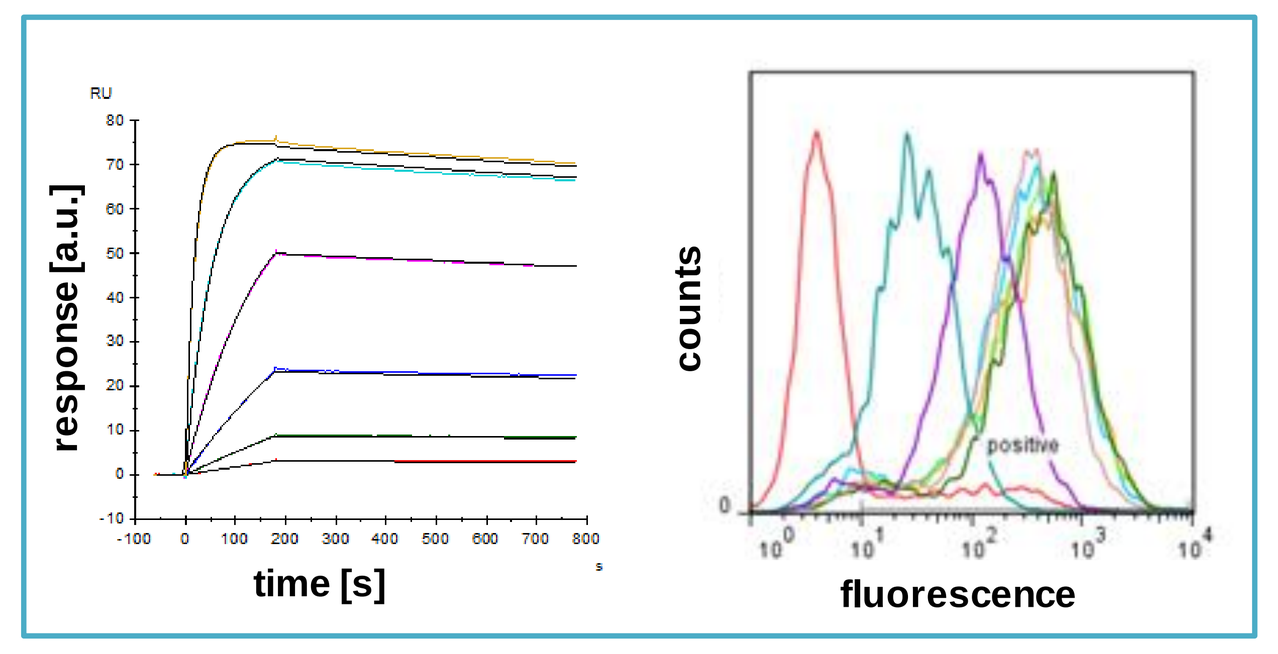
response [a.u.]
time [s]
counts
fluorescence

Figure 2. Characterization of CD137-binding Anticalin by SPR and FACS demonstrates high-affinity binding.

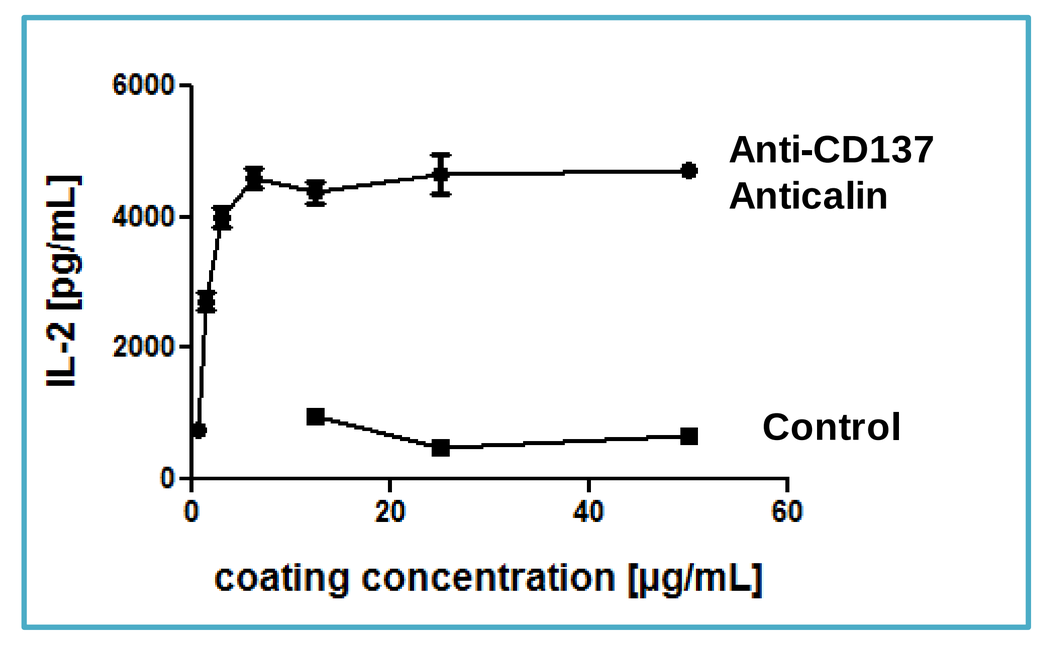
IL-2 [pg/mL]
Anti-CD137 Anticalin
Control
coating concentration [µg/mL]

Figure 3. Ex vivo assay demonstrates strong T cell costimulation (test articles coated together with sub-threshold concentrations of anti-CD3 antibody).

CD137/HER2 bispecifics: genetic fusion of anti-CD137 Anticalin with modified trastuzumab

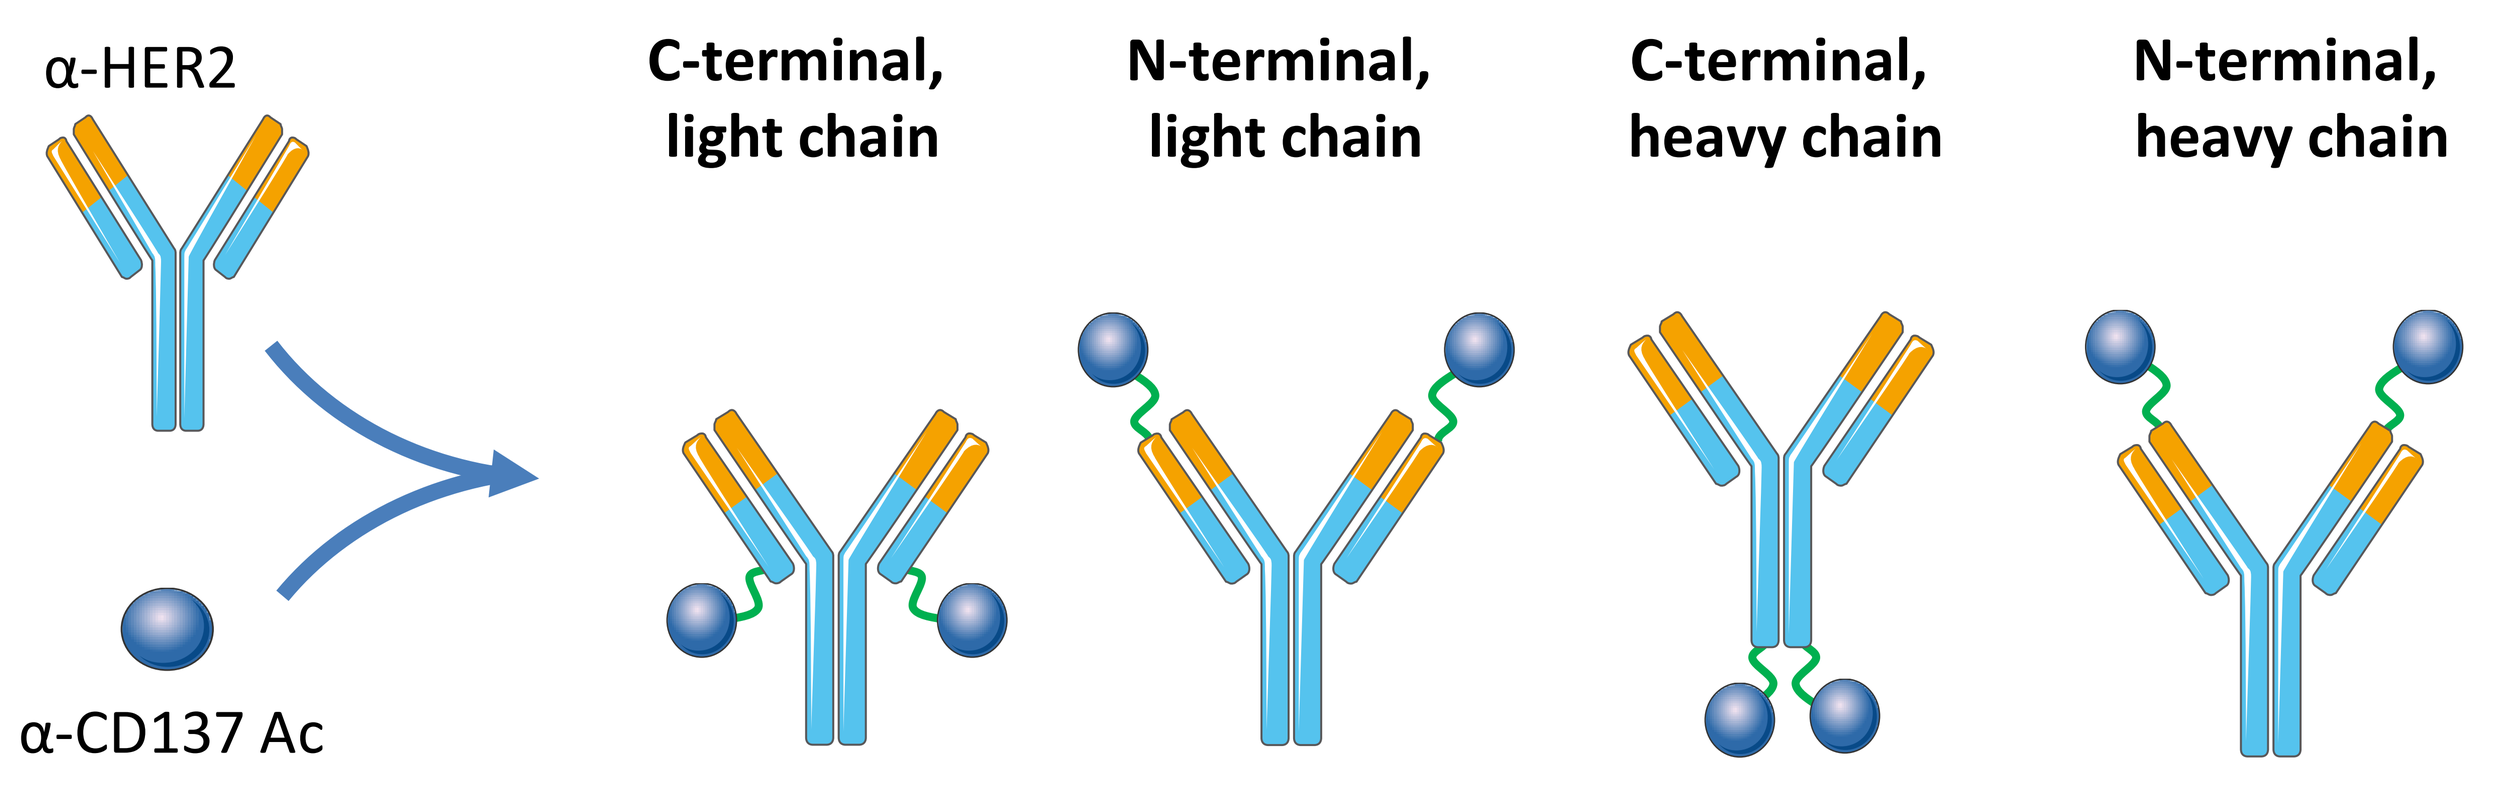
α-HER2
α-CD137 Ac
C-terminal, light chain
N-terminal, light chain
C-terminal, heavy chain
N-terminal, heavy chain

Figure 4. Design of four CD137/HER2 bispecifics: Genetic fusions of backbone-engineered trastuzumab to the anti-CD137 Anticalin were generated to either one of the four termini of the antibody. The IgG1 backbone of trastuzumab was exchanged for an engineered IgG4 backbone containing the stabilizing S228P mutation. Additional mutations to silence Fcγ-receptor interaction were made to exclude the possibility both of Fcγ-receptor-driven CD137 clustering, and antibody-dependent cell-mediated cytotoxicity (ADCC) against CD137-positive cells.

CD137/HER2 bispecifics are homogenous and stable

- Straightforward expression in CHO cells
- Fully stable and complete lack of aggregation after 1 week at 37°C in PBS
- Fully active after 1 week at 37°C in human and mouse plasma

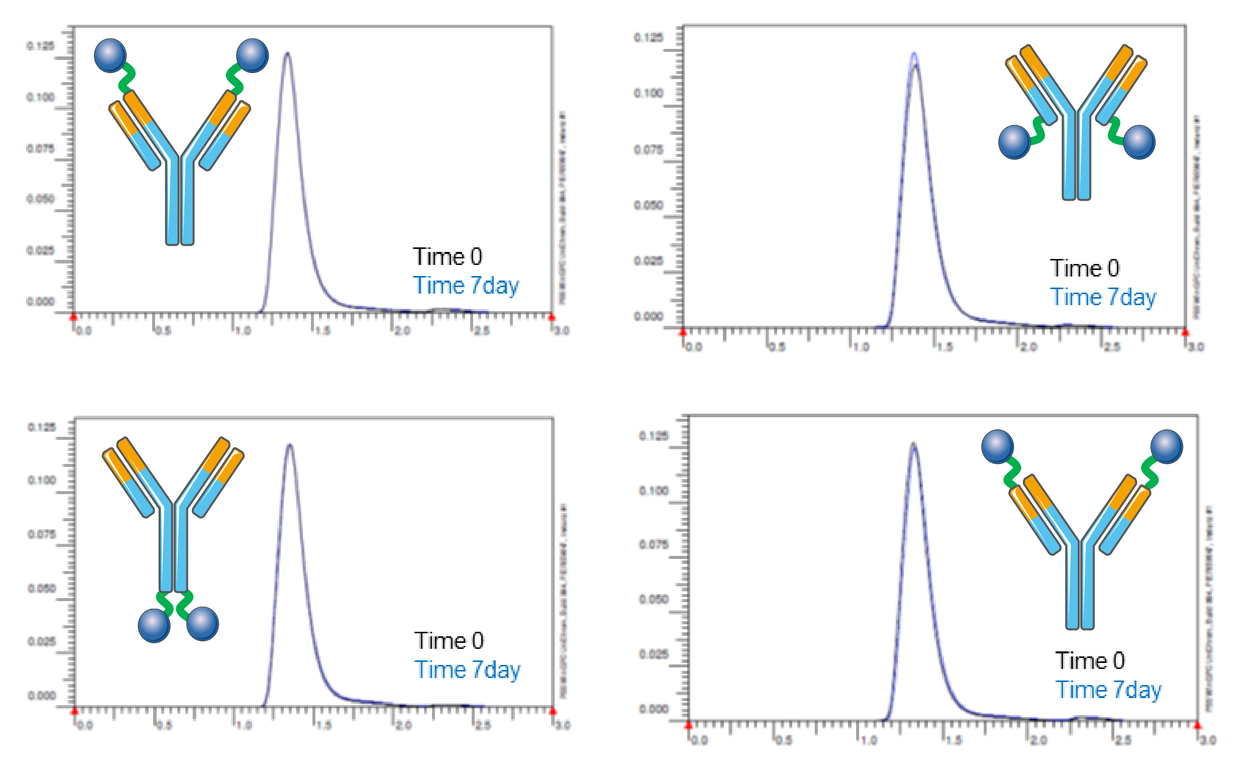
Time 0
Time 7day

Figure 5. CD137/HER2 bispecifics display a pristine SEC profile before and after 1 week at 37°C in PBS, pH7.4 (overlay).

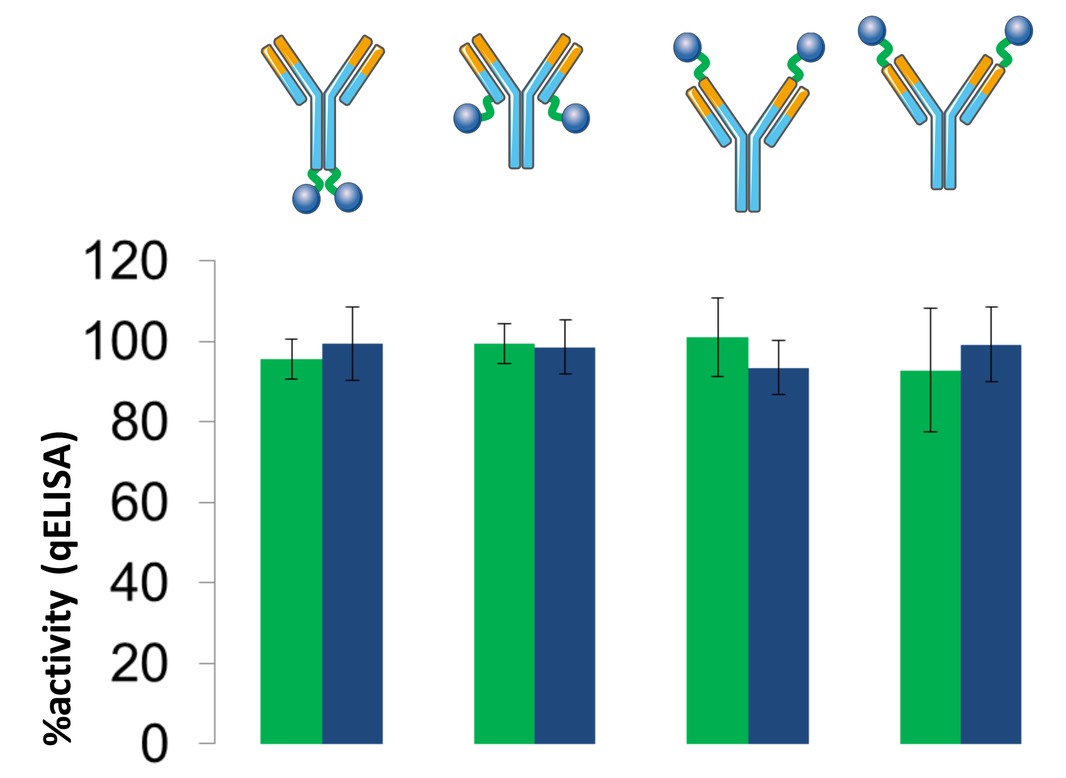
%activity (qELISA)

Figure 6. CD137/HER2 bispecifics remain fully active after 1 week at 37°C in human plasma (green) and mouse plasma (blue; quantitative ELISA).

Target binding: affinity of parental building blocks retained

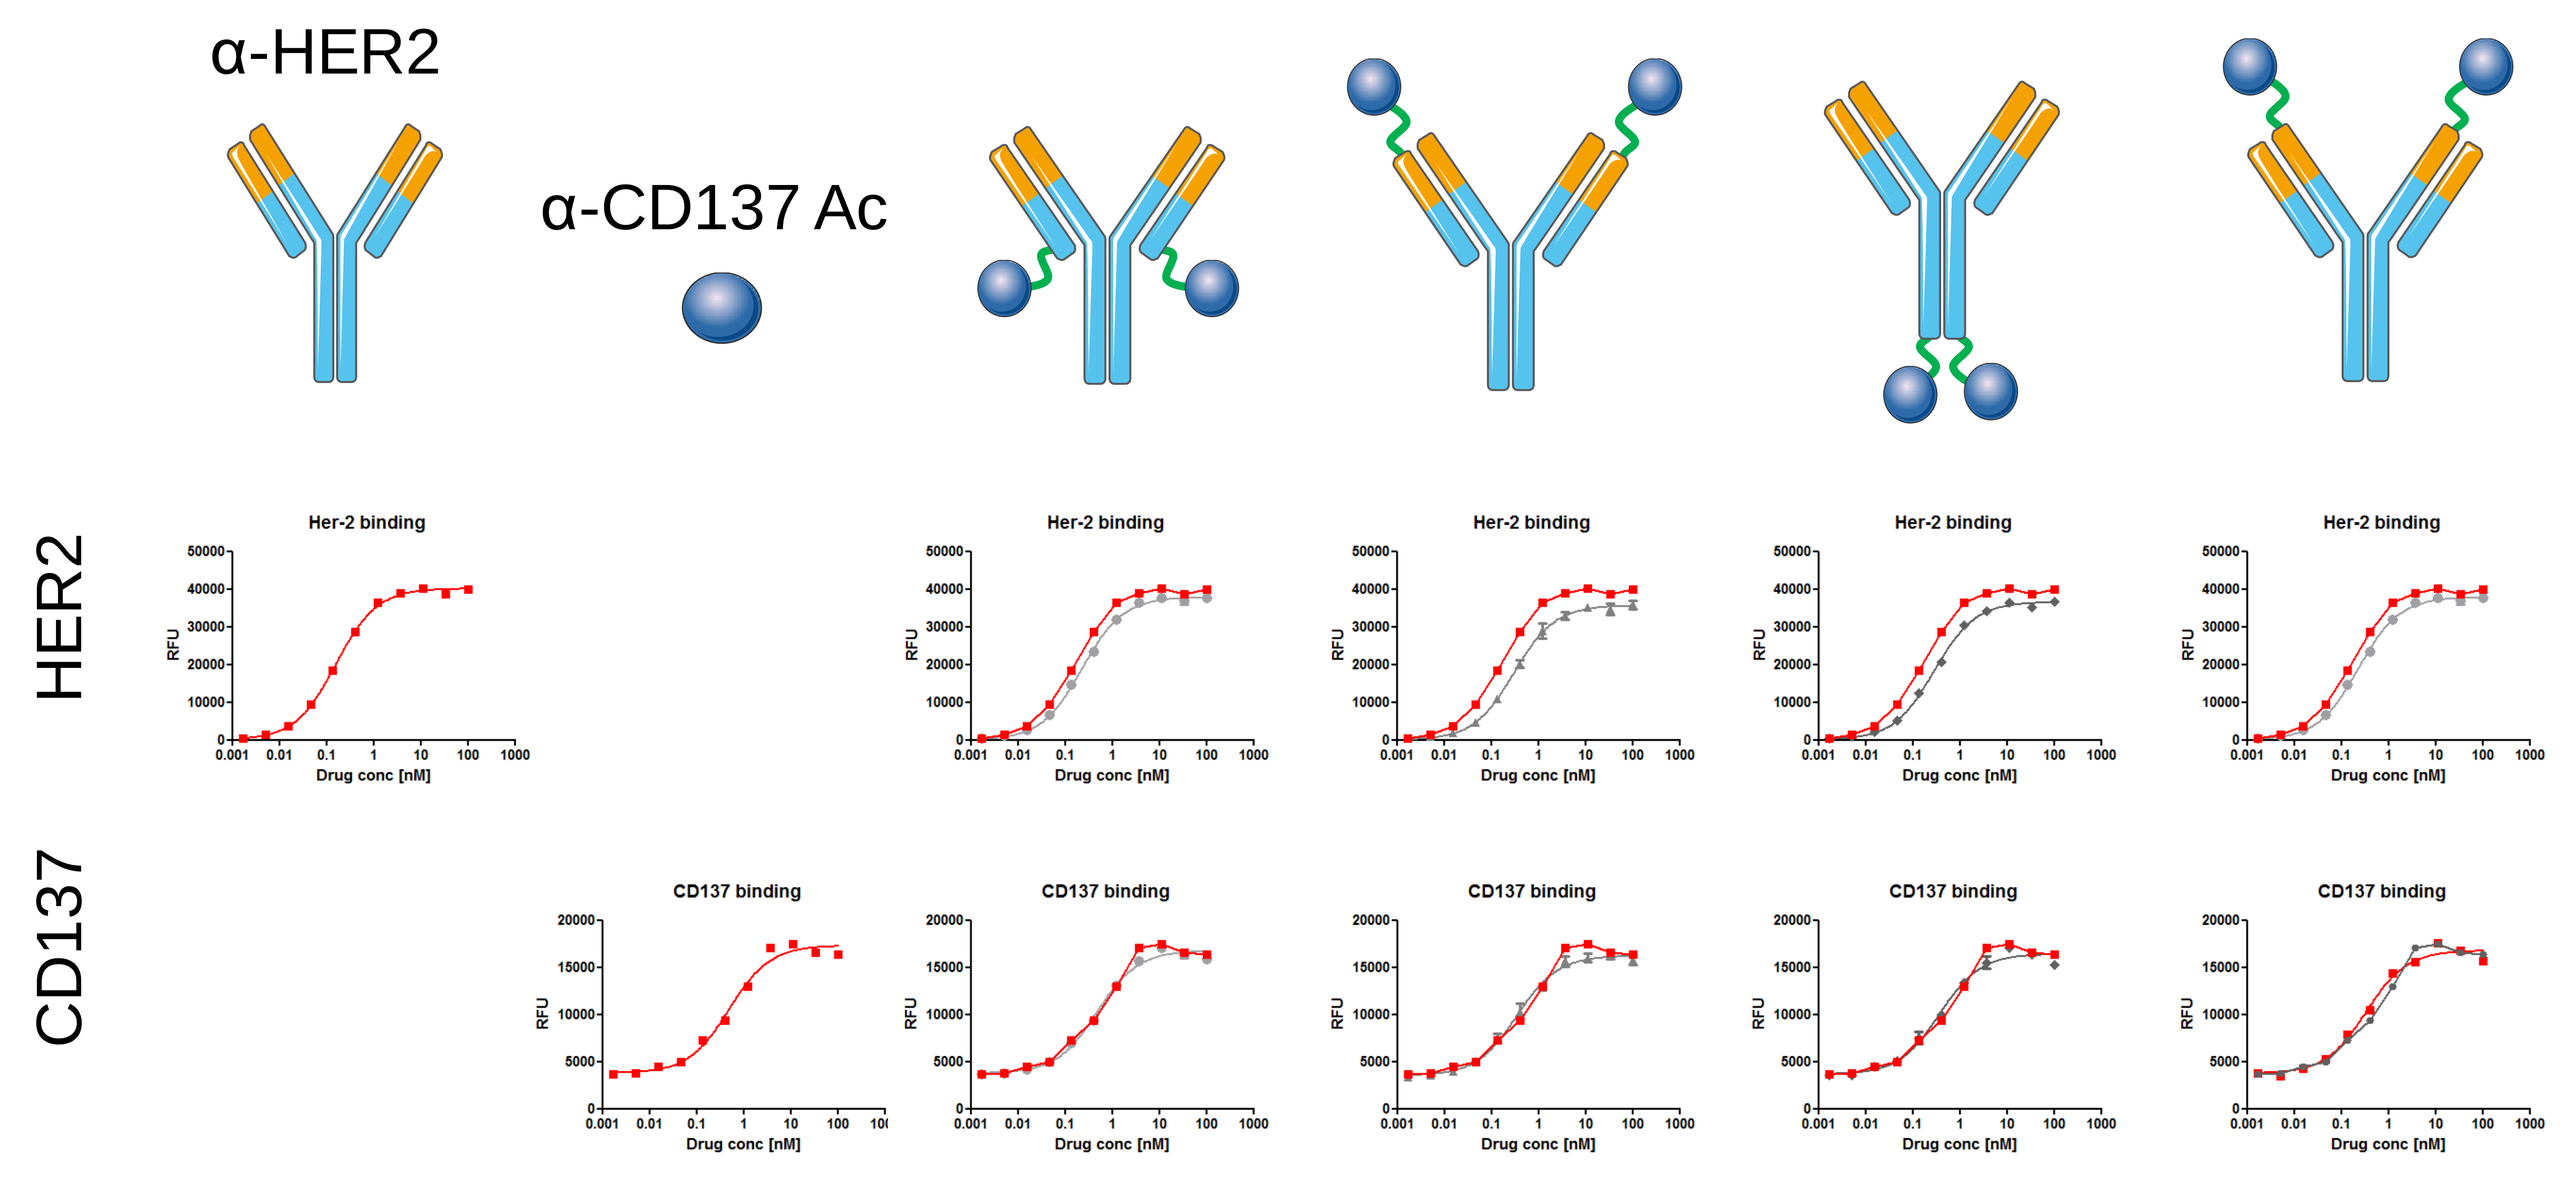
α-HER2
α-CD137 Ac
HER2
CD137
Her-2 binding
CD137 binding
Drug conc [nM]

Figure 7. ELISA: CD137/HER2 bispecifics bind both targets with identical affinity compared to the individual building blocks anti-CD137 Anticalin and trastuzumab.

Simultaneous binding of both targets confirmed for all CD137/HER2 bispecifics

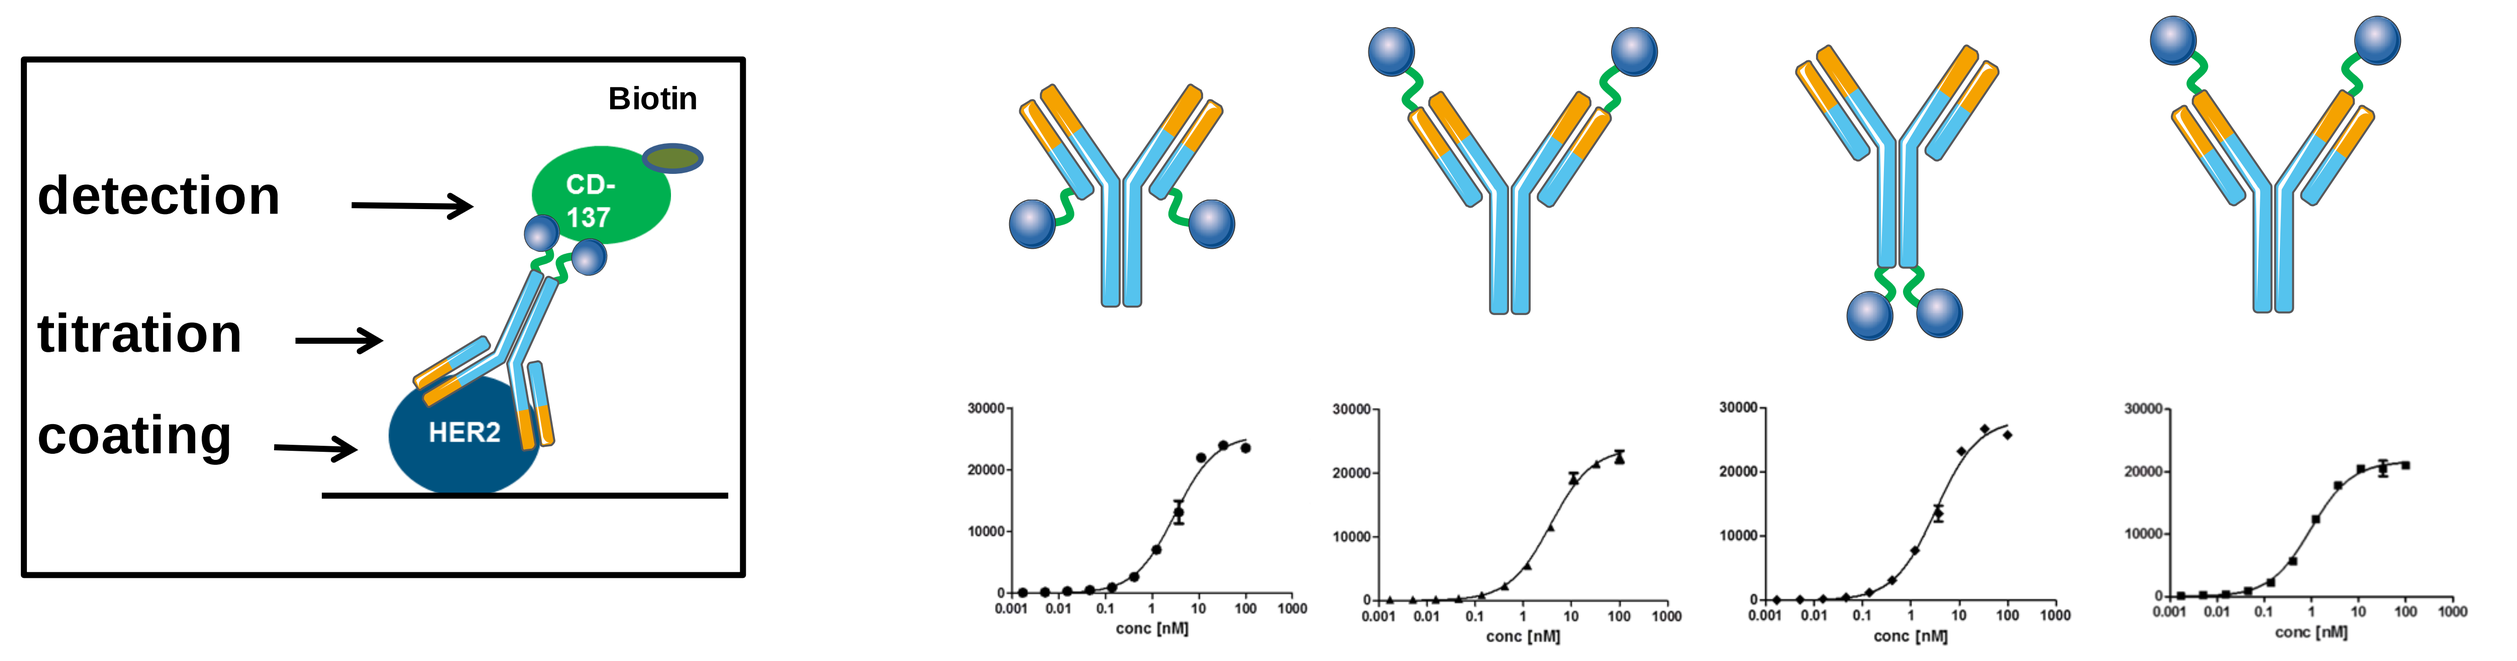
Biotin
detection
CD-137
titration
coating
HER2

Figure 8. Dual binding: CD137/HER2 bispecifics are capable to bind both targets at the same time according to Sandwich ELISA.

Engineered IgG4 backbone shows very low FcγR but retained FcRn binding

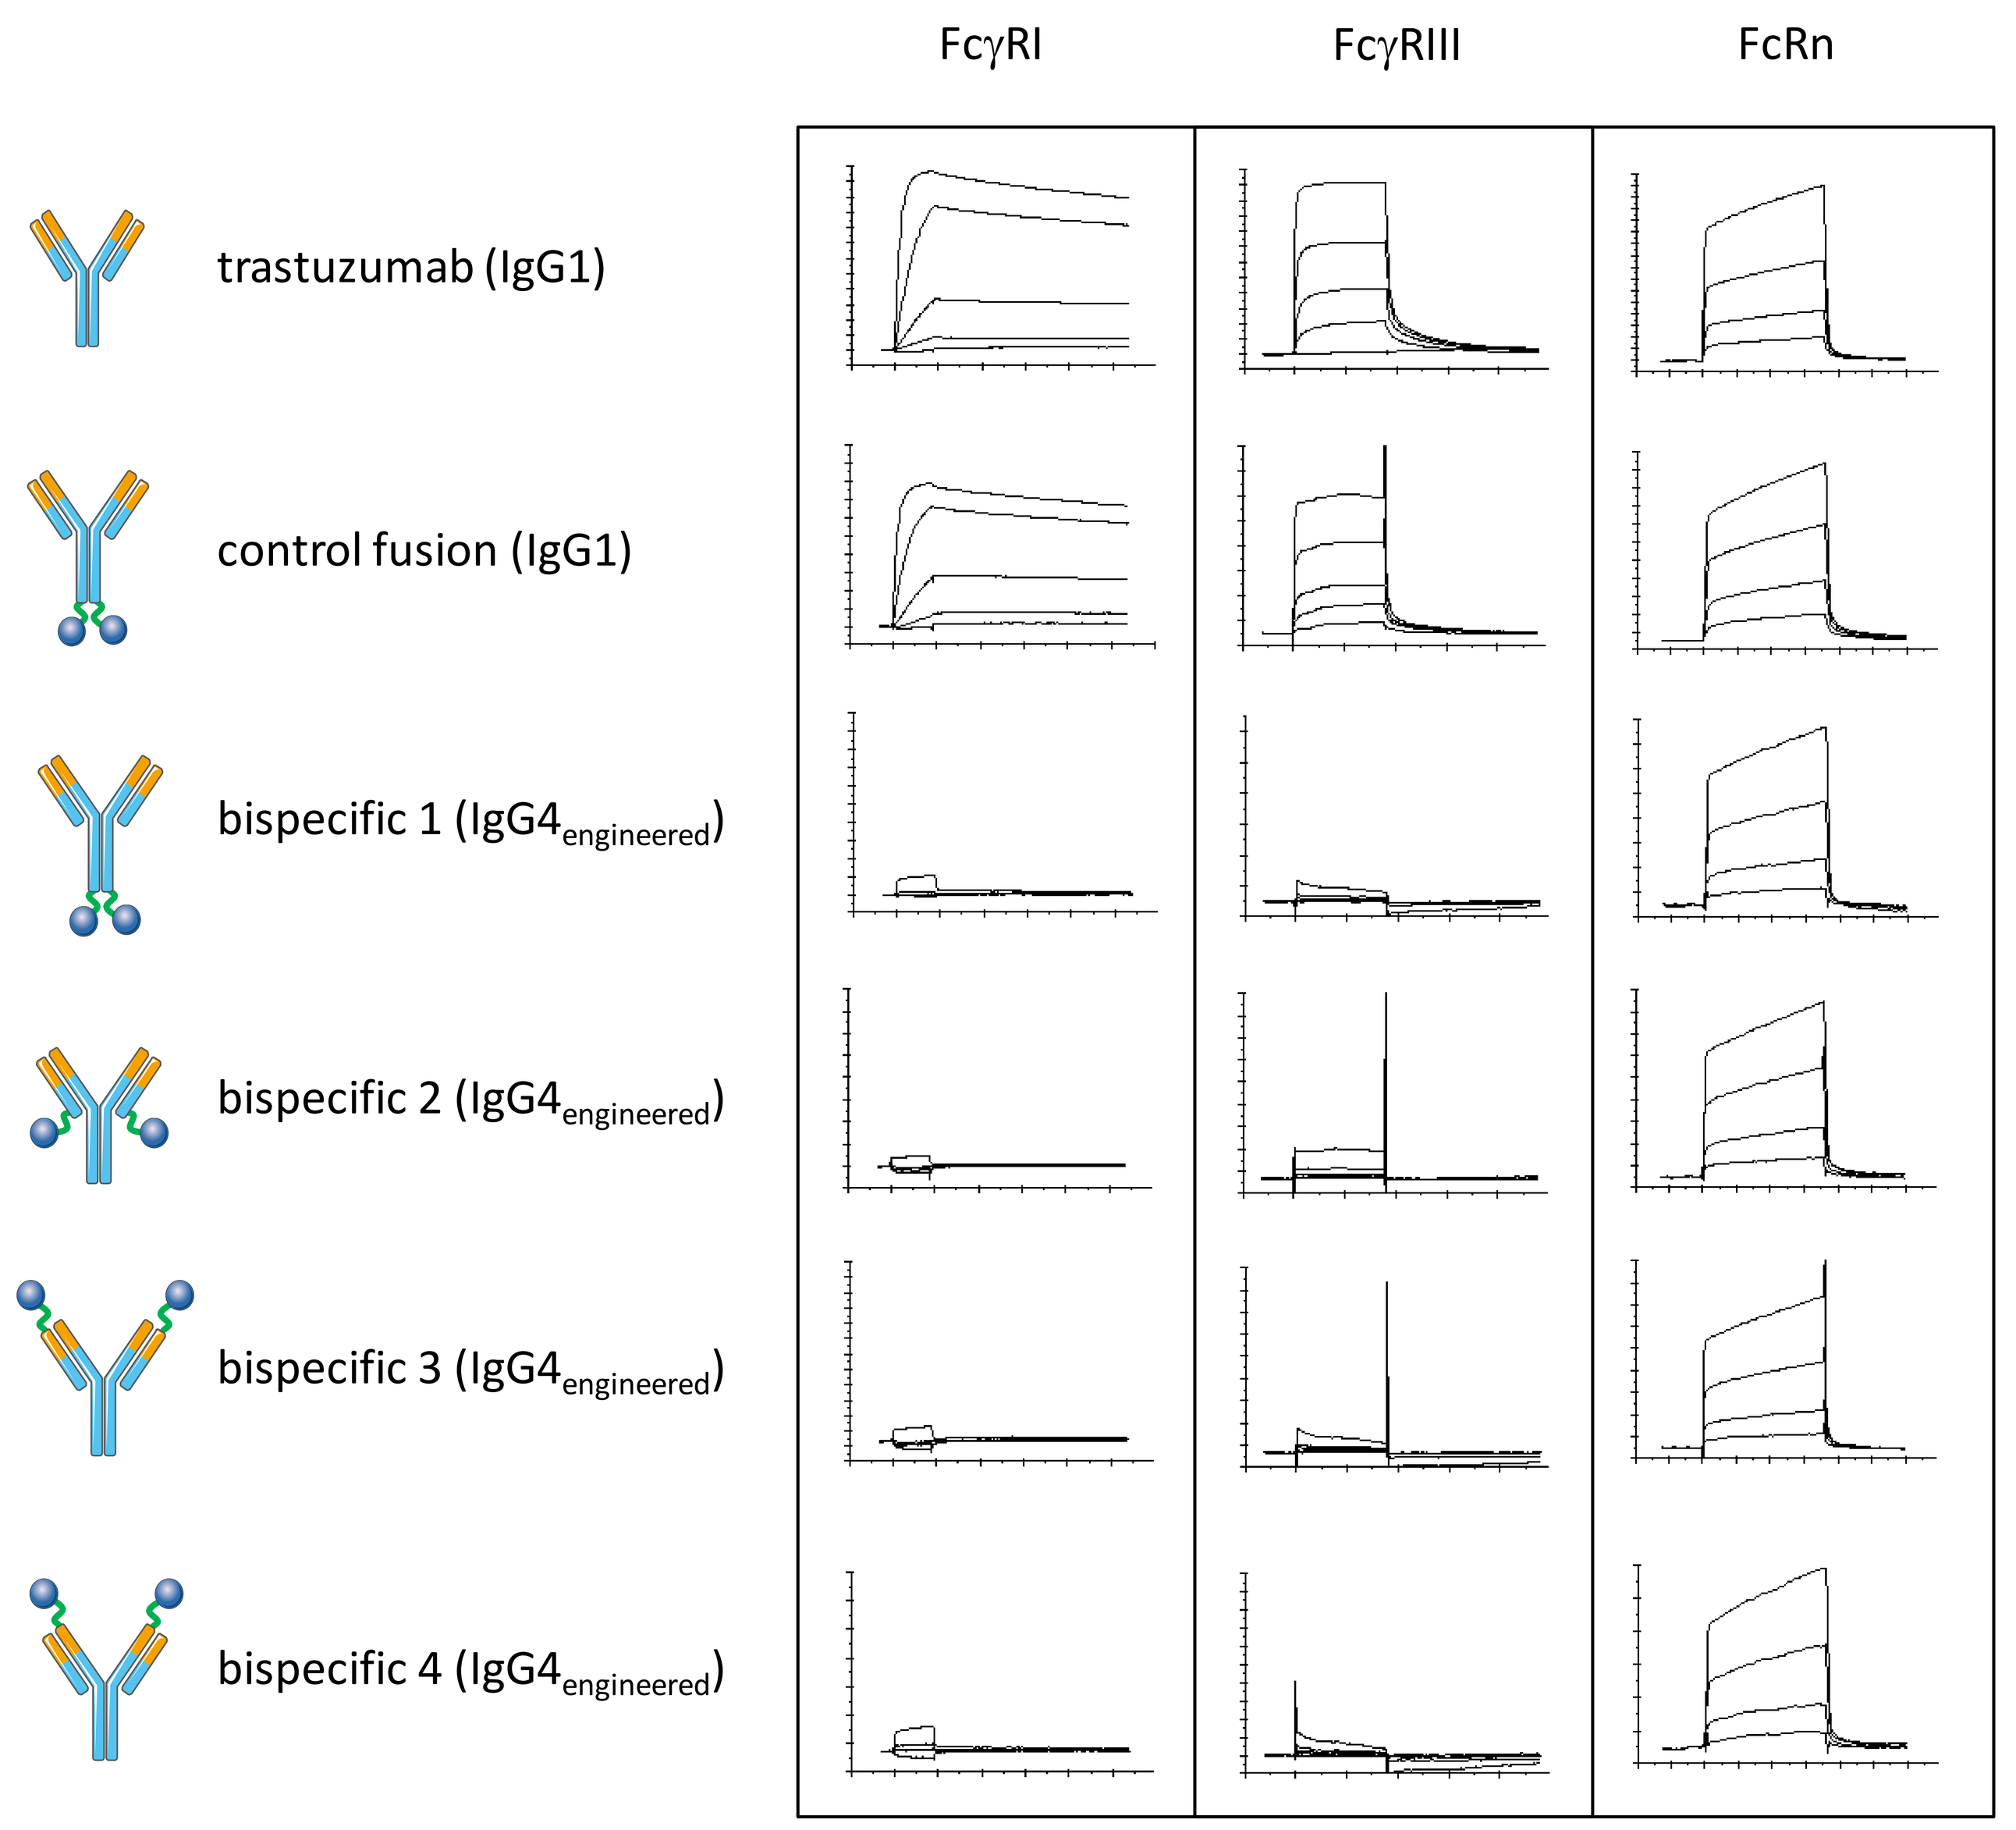
FcγRI
FcγRIII
FcRn
trastuzumab (IgG1)
control fusion (IgG1)
bispecific 1 (IgG4engineered)
bispecific 2 (IgG4engineered)
bispecific 3 (IgG4engineered)
bispecific 4 (IgG4engineered)

Figure 9. Surface plasmon resonance: Binding of CD137/HER2 bispecifics to recombinant FcγRI and FcγRIII is reduced compared to trastuzumab, while binding to the neonatal Fc receptor FcRn is retained.

Potent costimulatory T cell activation is dependent on both HER2 binding and bispecific geometry

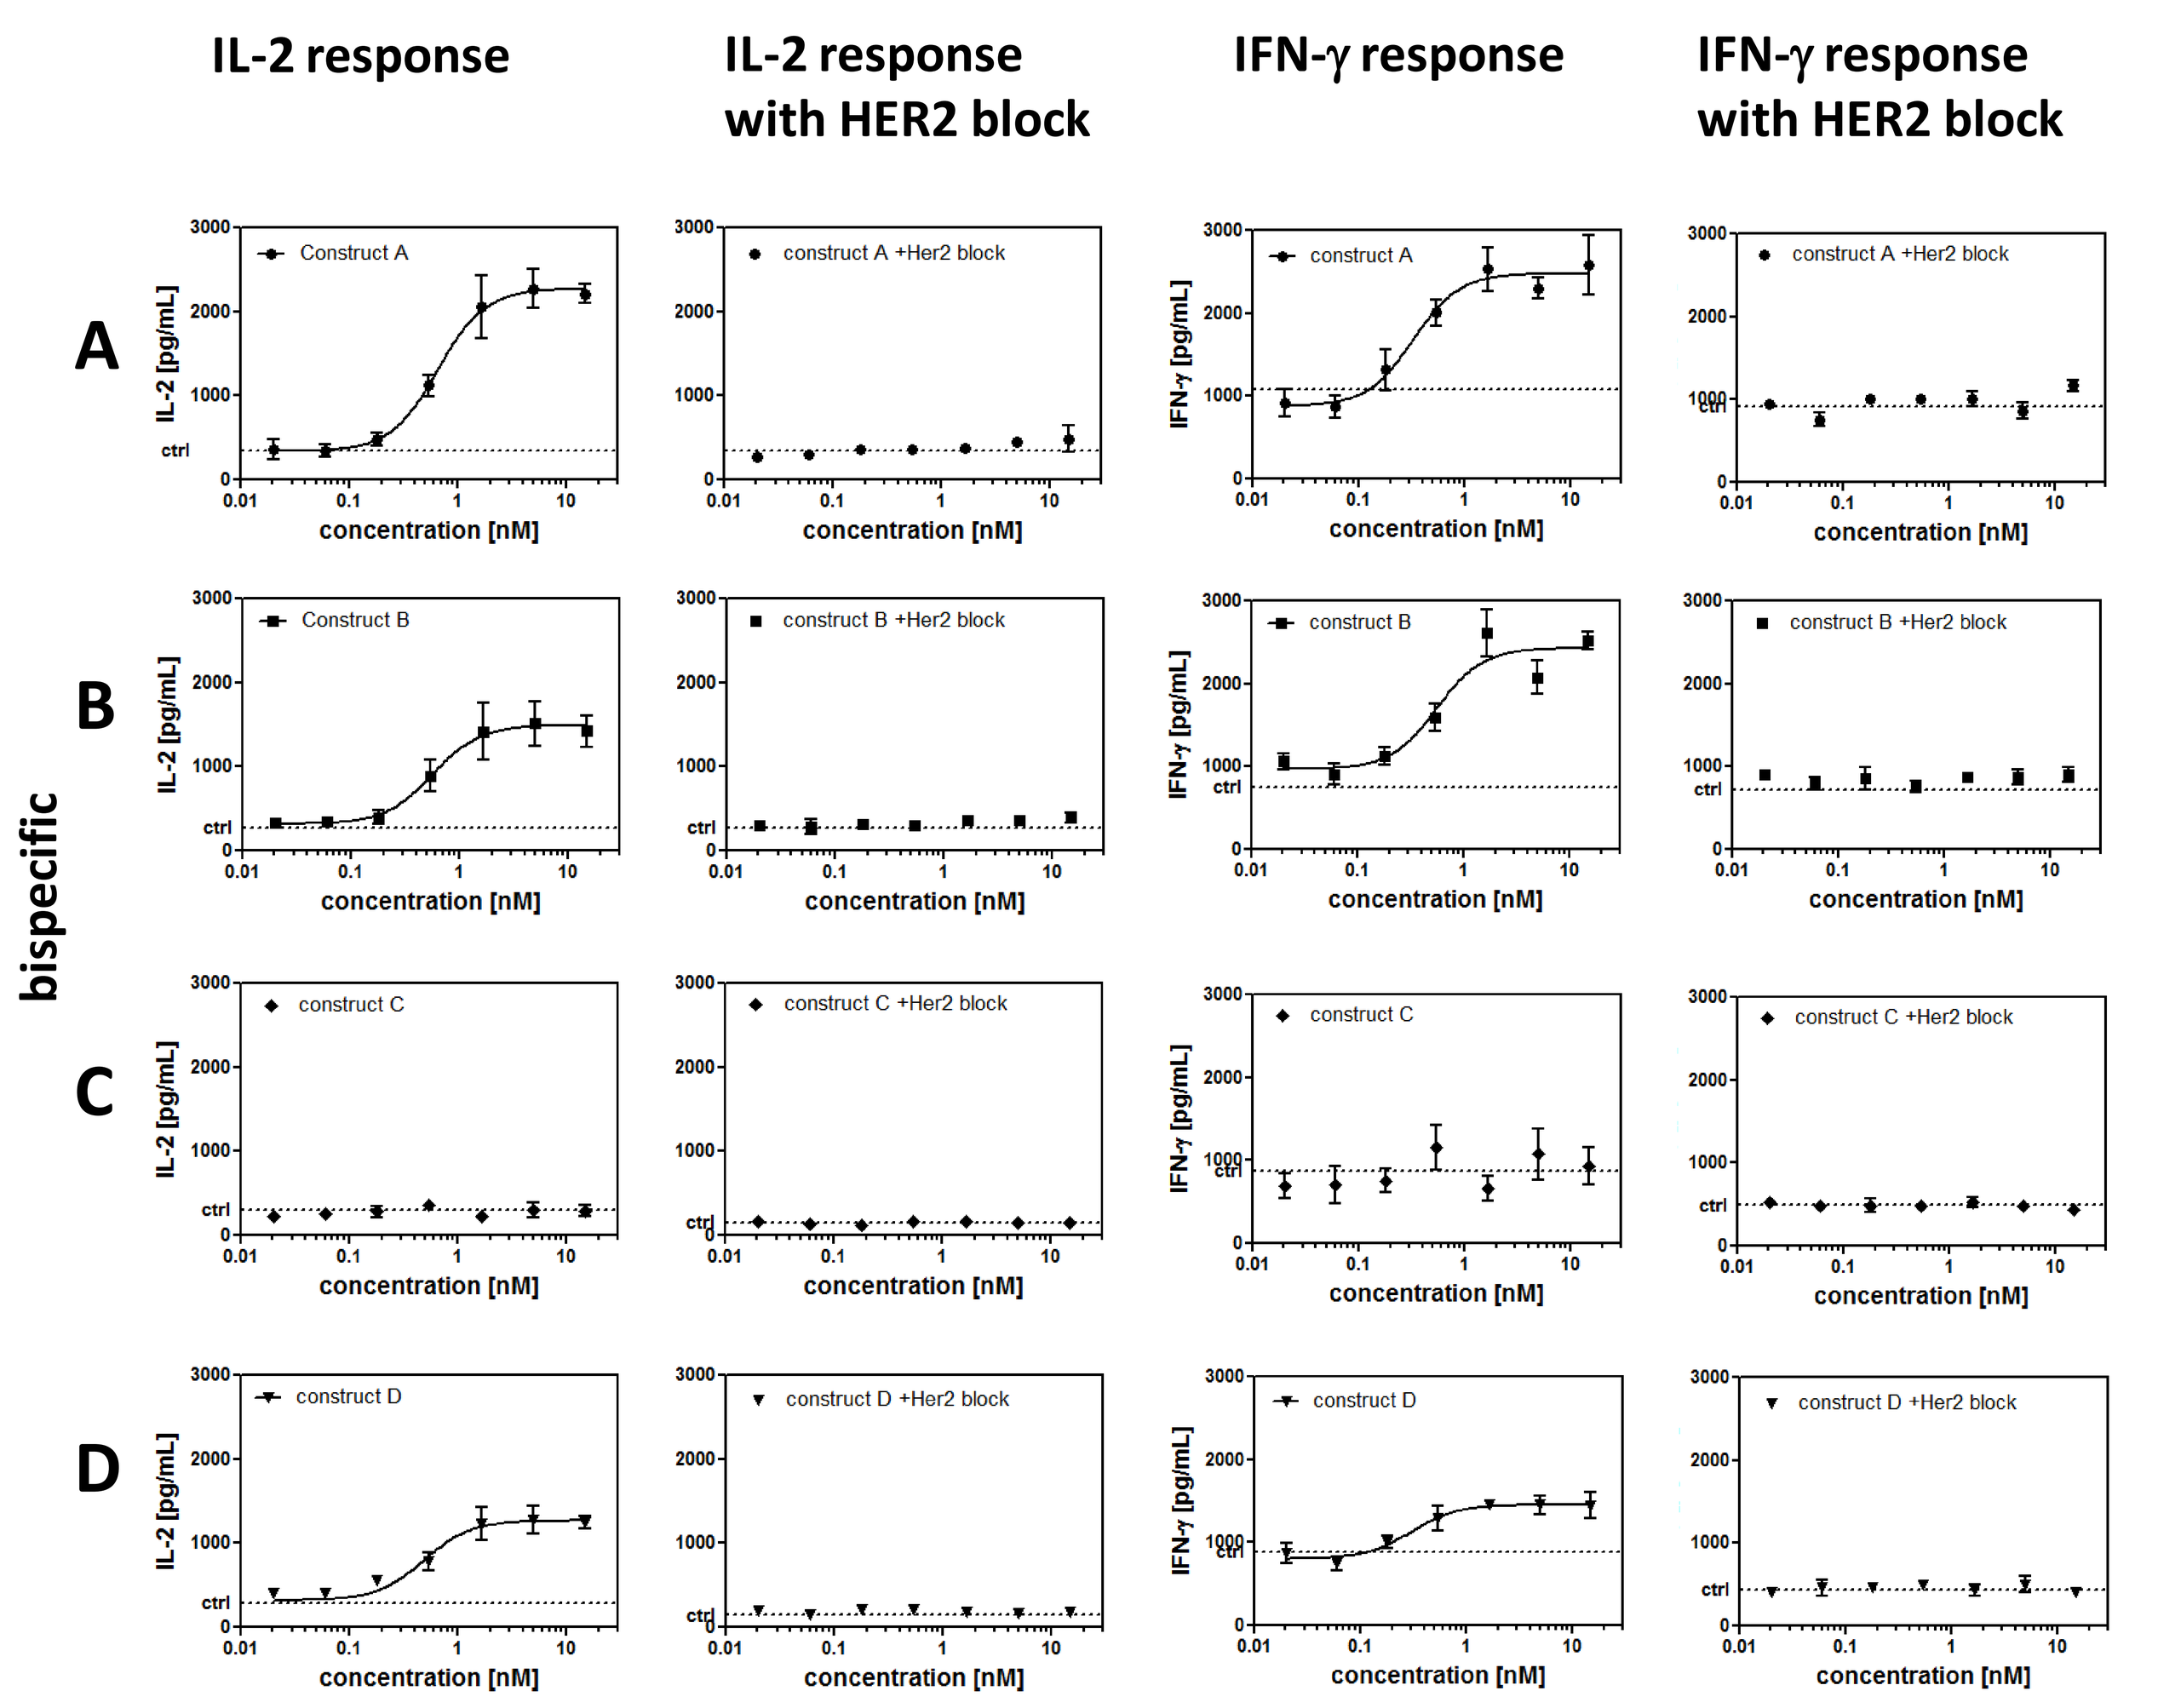
IL-2 response
IL-2 response with HER2 block
IFN-γ response
IFN-γ response with HER2 block
A
B
C
D
bispecific
IL-2 [pg/mL]
IFN-γ [pg/mL]
concentration [nM]

Figure 10. Ex vivo T cell activation: The four CD137/HER2 bispecifics A-D display different capabilities to elicit IL-2 and IFN-γ production by costimulatory engagement. In all cases the response is diminished to background levels by disrupting the binding of the bispecifics to cellular HER2 by adding an excess of trastuzumab.

Brief methods.
SKBR3 tumor cells were grown overnight on 96-well plates that had been precoated with anti-CD3 antibody. The next day T cells purified from healthy donor PBMC were added to the wells together with the titrated CD137/HER2 bispecific molecules (constructs A-D). After three days in culture, IL-2 and IFN-γ levels in the culture supernatants were measured by an Electrochemoluminescence immunoassay. The experiment was performed also in the presence of an excess of trastuzumab to inhibit the binding of CD137/HER2 bispecifics to the SKBR3 cells.

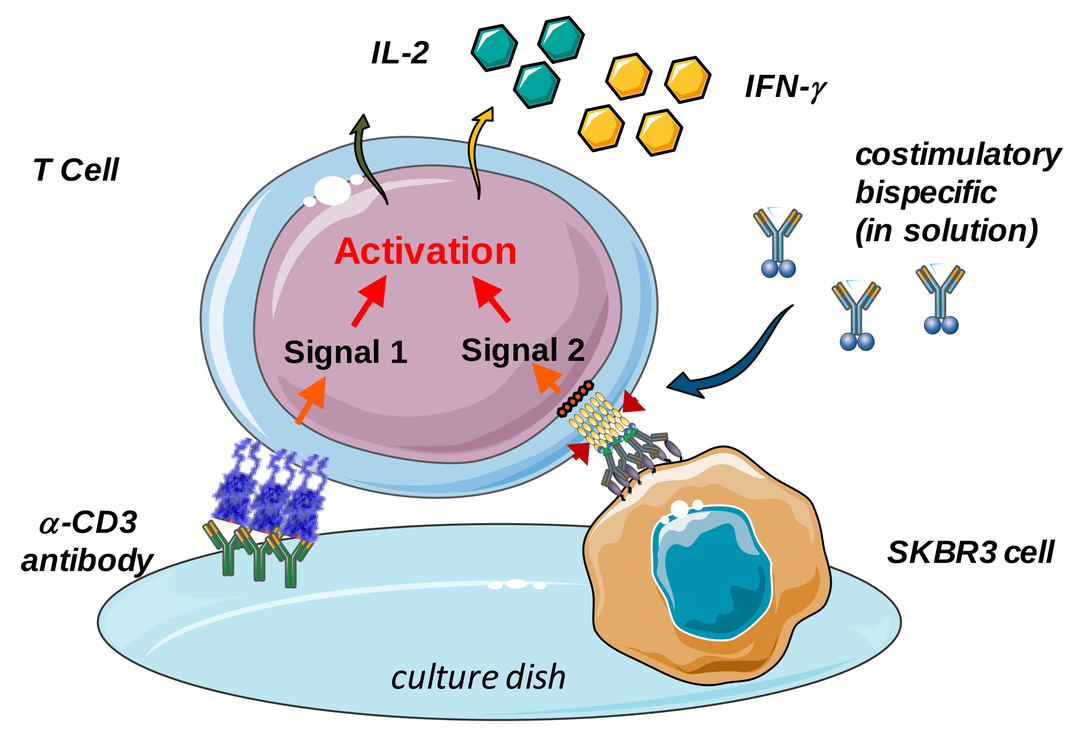
IL-2
IFN-γ
T Cell
costimulatory bispecific (in solution)
Activation
Signal 1
Signal 2
α-CD3 antibody
SKBR3 cell
culture dish

Summary

- **Four CD137/HER2 bispecifics were generated based on genetic fusion of high-affinity CD137-binding Anticalin and modified trastuzumab, displaying**
 - **excellent drug-like properties**
 - **simultaneous, dual target binding**
 - **maintained target affinity compared to parental building blocks**
 - **low FcγR-interaction of engineered backbone to exclude non-tumor-target driven clustering and ADCC against CD137-positive cells**
- **CD137/HER2 bispecifics induce strong T cell activation via tumor target-dependent costimulatory T cell engagement**
- **We expect the approach to allow potent local activation of tumor-specific T cells with manageable systemic toxicity**

References.
[1] Ye, Q. et al., Clin Canc Res 2014 Jan 1; 20(1):44-55. [2] Chacon, J. A. et al., PloS One 2013 8(4):e60031. [3] Kalos, M. et al., Sci Transl Med 2011 Aug 10; 3(95): 2-21. [4] Maude, S. L. et al., N Engl J Med 2014 Oct 16; 371(16):1507 – 1517. [5] Kohrt, H. et al, J Clin Invest. 2012 Mar;122(3):1066-75.[6] Palazon, A. et al., Cancer Discovery: 2012 (2): 608 – 623. [7] Pastor F. et al., Mol Ther. 2011 Oct;19(10):1878-86. [8] Mukai, Y. et al., Sci Signal.: 2010 (3): ra83.
Parts of images on this poster based on material from Servier Medical Art under a Creative Commons Attribution 3.0 unported license.